ABSTRACT 334

Plasma Cytokine and Angiogenic Factors Predictive of Clinical Benefit and Prognosis in Patients With Advanced or Metastatic Renal Cell Cancer Treated in Phase III Trials of Pazopanib

Yuan Liu[1], Hai T. Tran[2], Ying Lin[1], Anne-Marie Martin[1], Amado Zurita[2], Cora N. Sternberg[3], VEG105192 Team[4], Rafael Amado[1], Lini N. Pandite[1], John V. Heymach[2]

[1]GlaxoSmithKline, Research Triangle Park, NC, and Philadelphia, PA, USA; [2]University of Texas M.D. Anderson Cancer Center, Houston, TX, USA; [3]San Camillo Forlanini Hospitals, Department of Medical Oncology, Rome, Italy; [4]VEG105192 investigators, patients, and study team

BACKGROUND

- Renal cell carcinoma (RCC) is the most common cancer of the kidney
 - Recurrence develops in ~ 40% of patients with localized tumors and ~ 30% present with metastatic disease[1]
- Targeted agents have supplanted cytokines as the standard of care for the treatment of patients with advanced RCC[2]
- Pazopanib is an oral angiogenesis inhibitor targeting VEGFR-1, -2, and -3, PDGFR-α and -β, and c-Kit that has recently been granted broad regulatory approval for the treatment of patients with RCC[3-8]
- Few studies have analyzed the associations of baseline levels of molecular markers with disease prognosis and treatment outcomes of targeted agents; such analyses would facilitate individualized treatment decisions
- A previous exploratory analysis of a panel of 17 cytokines and angiogenic factors (CAFs) in patients with RCC treated with pazopanib demonstrated significant associations of baseline levels of 7 CAFs with therapeutic outcome and/or tumor burden[9]

OBJECTIVES

- To validate and extend findings of RCC phase II CAF studies on associations of baseline levels of select CAFs with: 1) disease prognosis in placebo-treated patients and 2) treatment outcome in pazopanib-treated patients with advanced RCC

METHODS

- Baseline plasma levels of select candidate CAFs identified in a previous study[9] were measured in a set of patients with adequate plasma samples who had participated in a randomized, placebo-controlled phase III trial (VEG105192)
 - Intent-to-treat population: N = 435; patients with plasma samples: n = 360
 - Plasma levels were quantitated using the SearchLight Multiplex Platform in a CLIA-certified laboratory (Aushon Biosystems, MA) (**Figure 1**)
- Biomarker population analyzed in the current study (n = 344)
 - Included 225 pazopanib-treated patients and 119 placebo-treated patients
 - Baseline and disease characteristics in these patient subsets were similar to the original study
- Marker levels were correlated with progression-free survival (PFS) by Cox regression and their Kaplan-Meier curves were plotted using median concentration of each marker as a cutoff
- Baseline levels of CAFs were defined as prognostic markers if they correlated with overall disease outcome, irrespective of treatment
 - Prognostic makers were therefore analyzed using placebo-treated patients
- Baseline levels of CAFs were defined as predictive markers if they correlated with treatment effects on efficacy endpoints
 - Predictive markers were therefore analyzed using both pazopanib- and placebo-treated patients

Figure 1. Study Flow

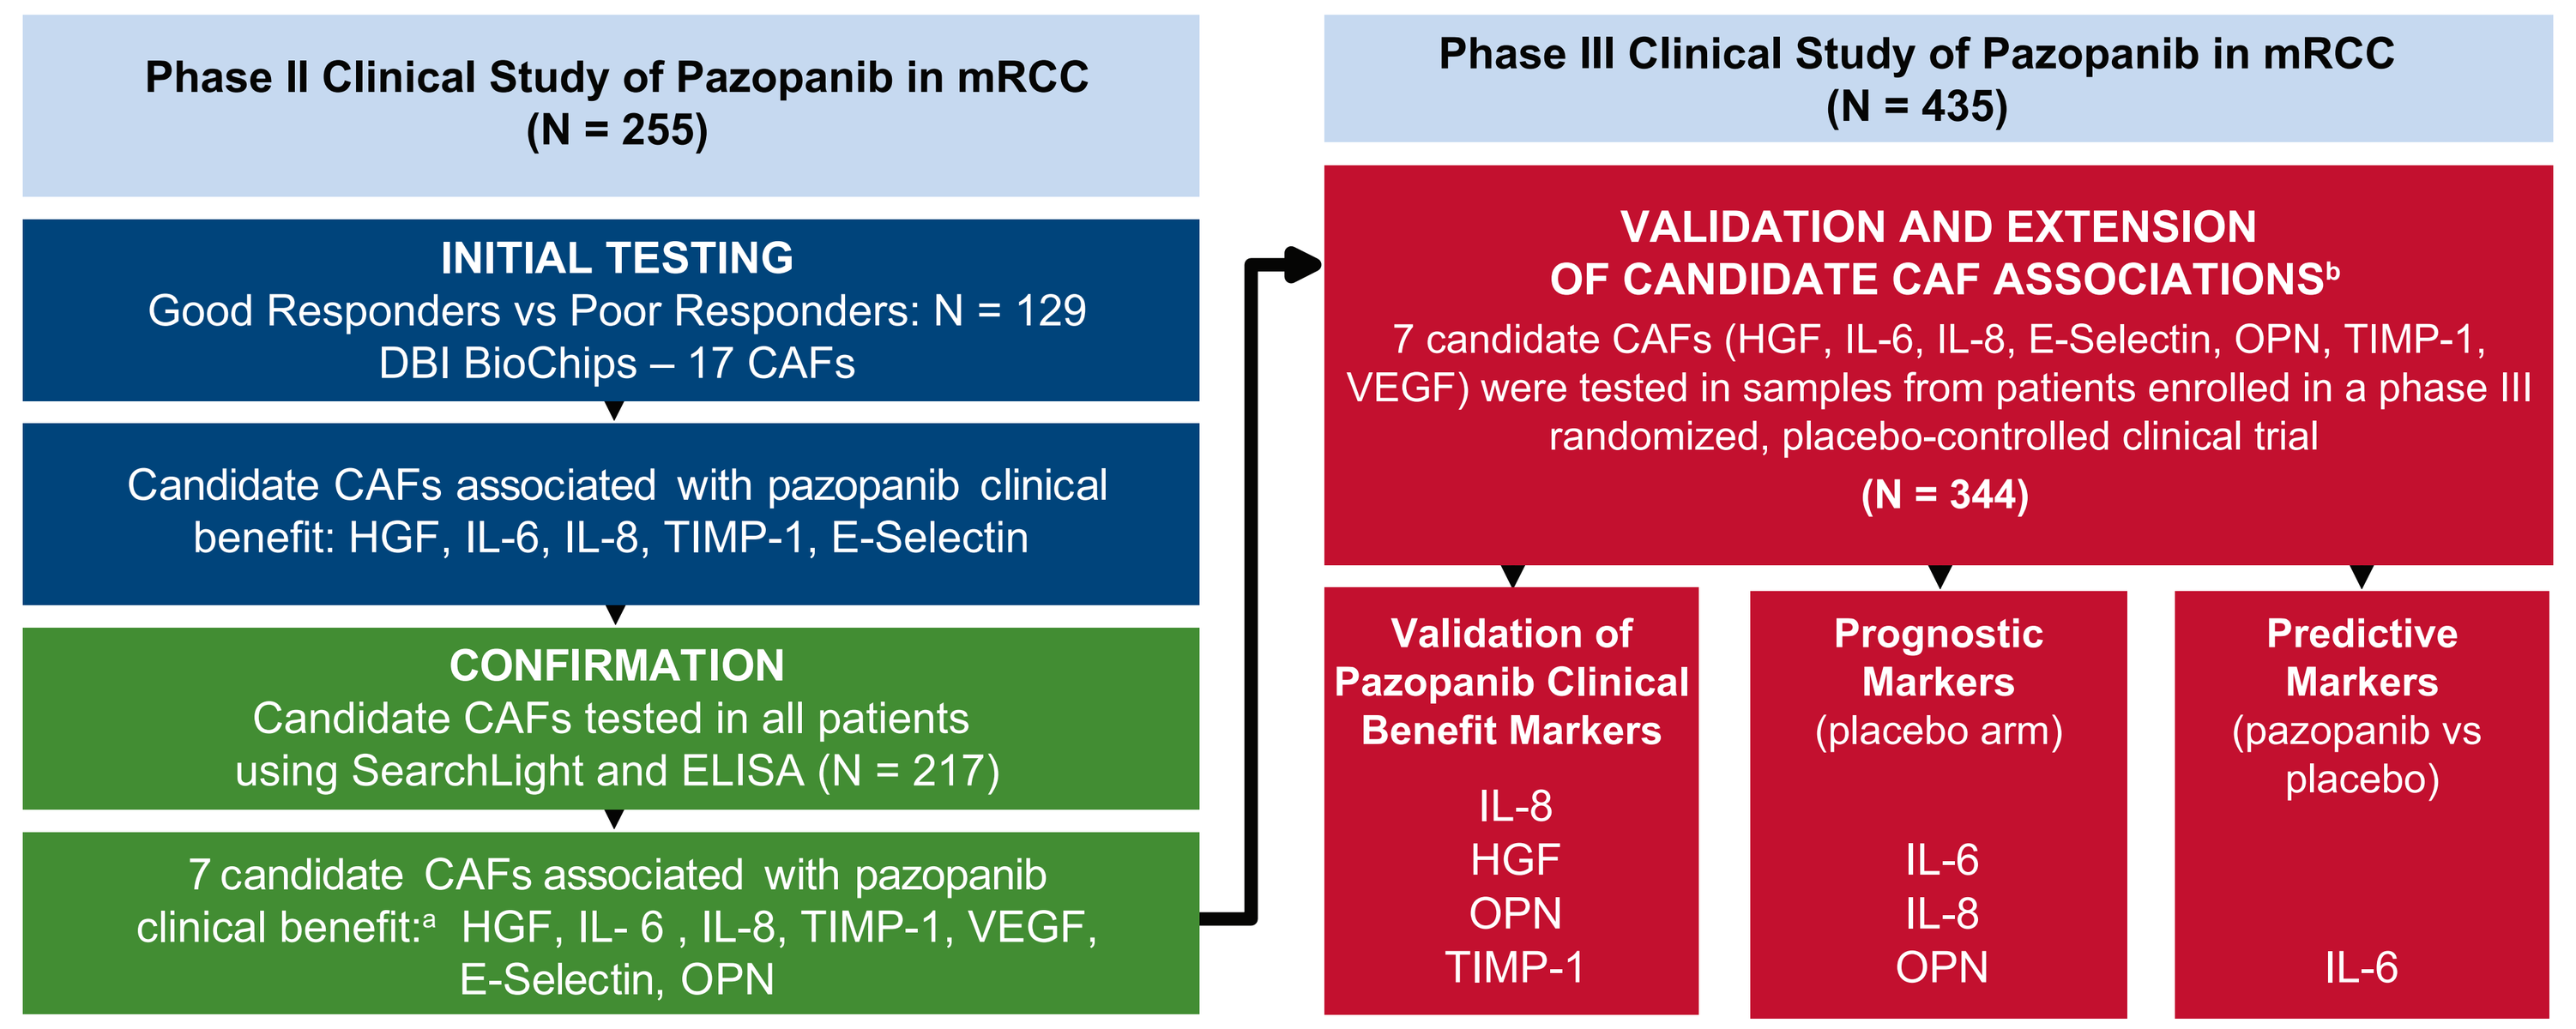
Phase II Clinical Study of Pazopanib in mRCC (N = 255)
INITIAL TESTING
Good Responders vs Poor Responders: N = 129
DBI BioChips – 17 CAFs
Candidate CAFs associated with pazopanib clinical benefit: HGF, IL-6, IL-8, TIMP-1, E-Selectin
CONFIRMATION
Candidate CAFs tested in all patients using SearchLight and ELISA (N = 217)
7 candidate CAFs associated with pazopanib clinical benefit:ᵃ HGF, IL-6, IL-8, TIMP-1, VEGF, E-Selectin, OPN
Phase III Clinical Study of Pazopanib in mRCC (N = 435)
VALIDATION AND EXTENSION OF CANDIDATE CAF ASSOCIATIONSᵇ
7 candidate CAFs (HGF, IL-6, IL-8, E-Selectin, OPN, TIMP-1, VEGF) were tested in samples from patients enrolled in a phase III randomized, placebo-controlled clinical trial
(N = 344)
Validation of Pazopanib Clinical Benefit Markers
IL-8
HGF
OPN
TIMP-1
Prognostic Markers (placebo arm)
IL-6
IL-8
OPN
Predictive Markers (pazopanib vs placebo)
IL-6

[a] Endpoints used for clinical benefit correlations included tumor response and progression-free survival.
[b] The focus of the current analysis is the Validation and Extension of Candidate CAF Associations.

Table 1. Characteristics of Patients in Biomarker Substudy

Parameter	Pazopanib (n = 225)	Placebo (n = 119)
Age, median years (range)	59 (31 - 85)	60 (25 - 81)
Sex, male, n (%)	149 (66)	89 (75)
Race, n (%)		
White	195 (87)	98 (82)
Asian	28 (12)	21 (18)
Other	2 (1)	0 (0)
ECOG performance status, n (%)		
0	93 (41)	45 (38)
1	132 (59)	74 (62)
MSKCC risk category		
Favorable risk	88 (39)	46 (39)
Intermediate risk	124 (55)	63 (53)
Poor risk	7 (3)	5 (4)
Unknown	6 (3)	5 (4)
Prior nephrectomy	198 (88)	104 (87)
Prior cytokine treatment	119 (53)	60 (50)

Abbreviations: ECOG, Eastern Cooperative Oncology Group; MSKCC, Memorial Sloan-Kettering Cancer Center.

RESULTS

Table 2. Validation of Pazopanib Clinical Benefit Markers (Pazopanib Arm Only)

CAF	Group	Median PFS, weeks	HR	95% CI	*P* Value
IL-8 (cutoff = 16.42 pg/mL)	Low	49.1	0.63	0.45 - 0.88	.006
	High	31.3			
	Total	39.6			
HGF (cutoff = 311.3 pg/mL)	Low	48.1	0.64	0.46 - 0.91	.01
	High	32.1			
	Total	39.6			
OPN (cutoff = 191.63 ng/mL)	Low	49.3	0.55	0.39 - 0.77	< .001
	High	32.0			
	Total	39.6			
TIMP-1 (cutoff = 676.07 ng/mL)	Low	49.1	0.63	0.45 - 0.88	.006
	High	32.6			
	Total	39.6			

Abbreviations: CI, confidence interval; HR, hazard ratio; PFS, progression-free survival.

Figure 2. Validation of CAF Markers of Clinical Benefit in the Pazopanib Arm. Panel A: Patients with low IL-8 have longer PFS than patients with high IL-8 (49.1 vs 31.3 weeks, respectively). Panel B: Patients with low HGF have longer PFS than patients with high HGF (48.1 vs 32.1 weeks, respectively). Panel C: Patients with low TIMP-1 have longer PFS than patients with high TIMP-1 (49.1 vs 32.6 weeks, respectively). Panel D: Patients with low OPN have longer PFS than patients with high OPN (49.3 vs 32.0 weeks, respectively).

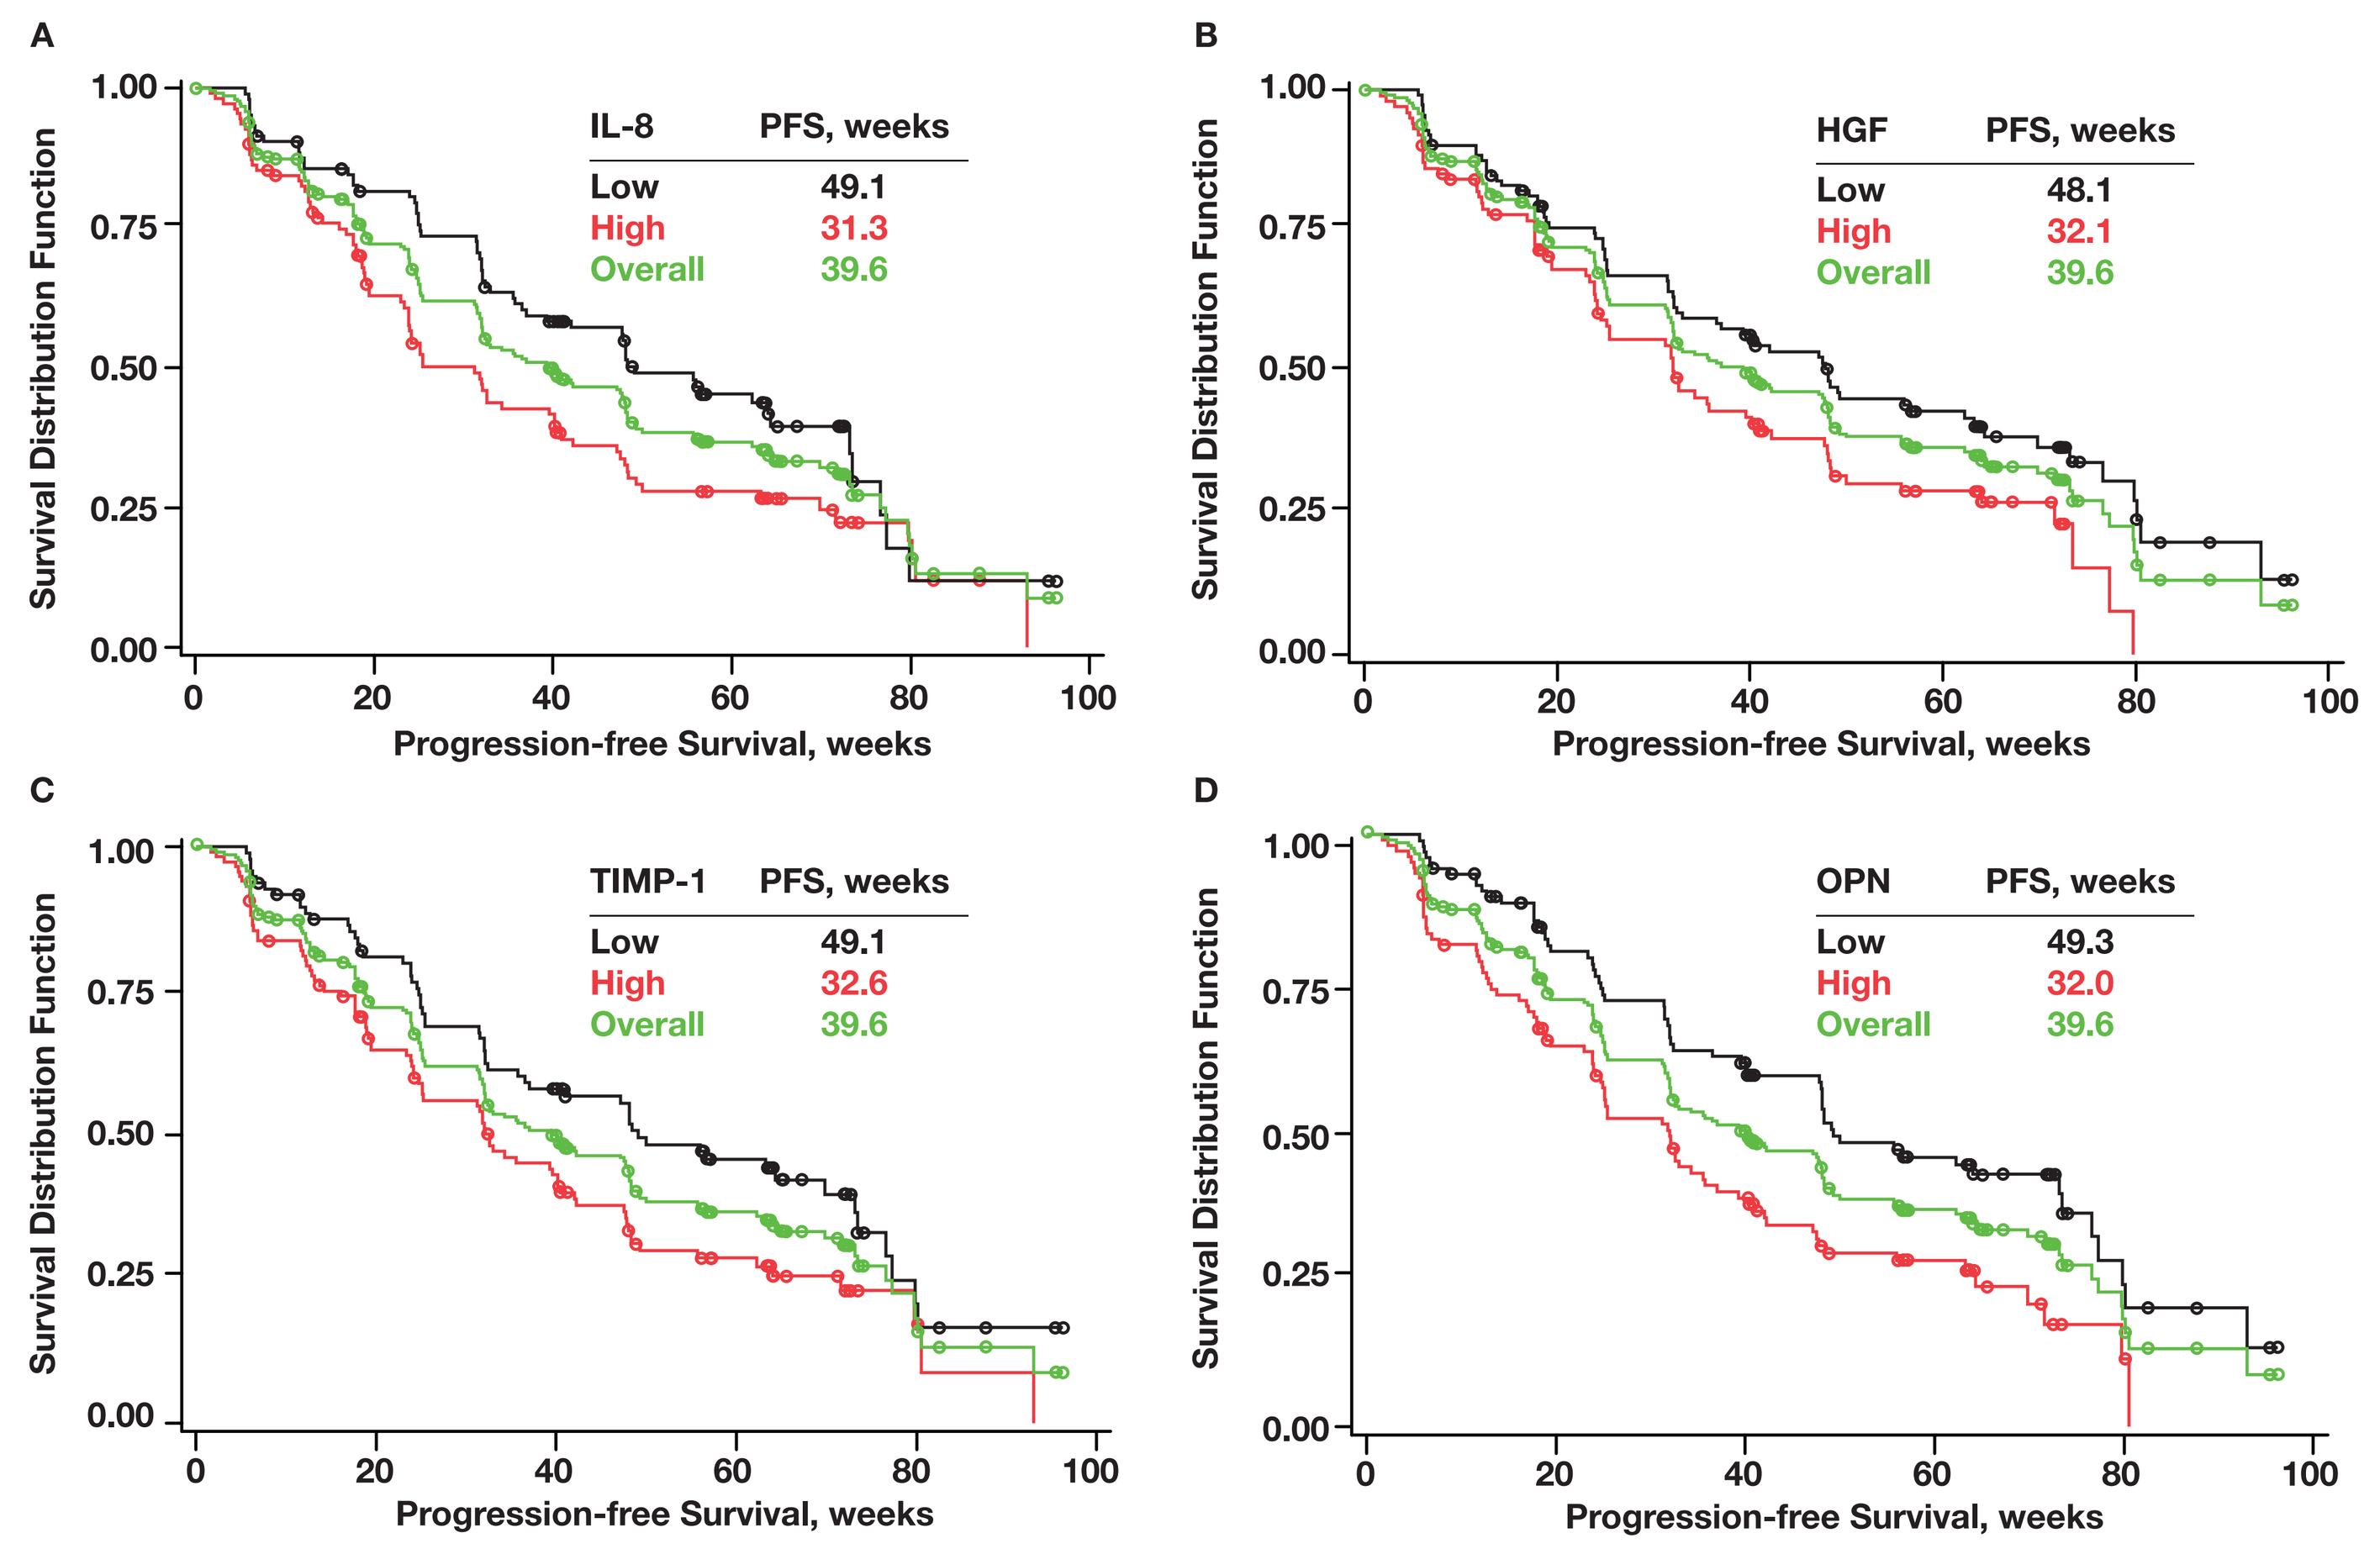
A
IL-8 PFS, weeks
Low 49.1
High 31.3
Overall 39.6
B
HGF PFS, weeks
Low 48.1
High 32.1
Overall 39.6
C
TIMP-1 PFS, weeks
Low 49.1
High 32.6
Overall 39.6
D
OPN PFS, weeks
Low 49.3
High 32.0
Overall 39.6
Survival Distribution Function
Progression-free Survival, weeks

Table 3. Prognostic Markers

CAF	Group	Median PFS, weeks	HR	95% CI	*P* (placebo)
IL-6	Low	24.0	0.458	0.31 - 0.68	< .001
	High	9.9			
	Total	13.4			
IL-8	Low	23.9	0.543	0.37 - 0.81	.002
	High	8.4			
	Total	13.4			
OPN	Low	24.0	0.437	0.29 - 0.66	< .001
	High	8.4			
	Total	13.4			
TIMP-1	Low	18.0	0.674	0.45 - 1.01	.052
	High	13.0			
	Total	14.1			

RESULTS (continued)

Figure 3. OPN and IL-8 PFS Analysis Using Baseline Median Cutoff

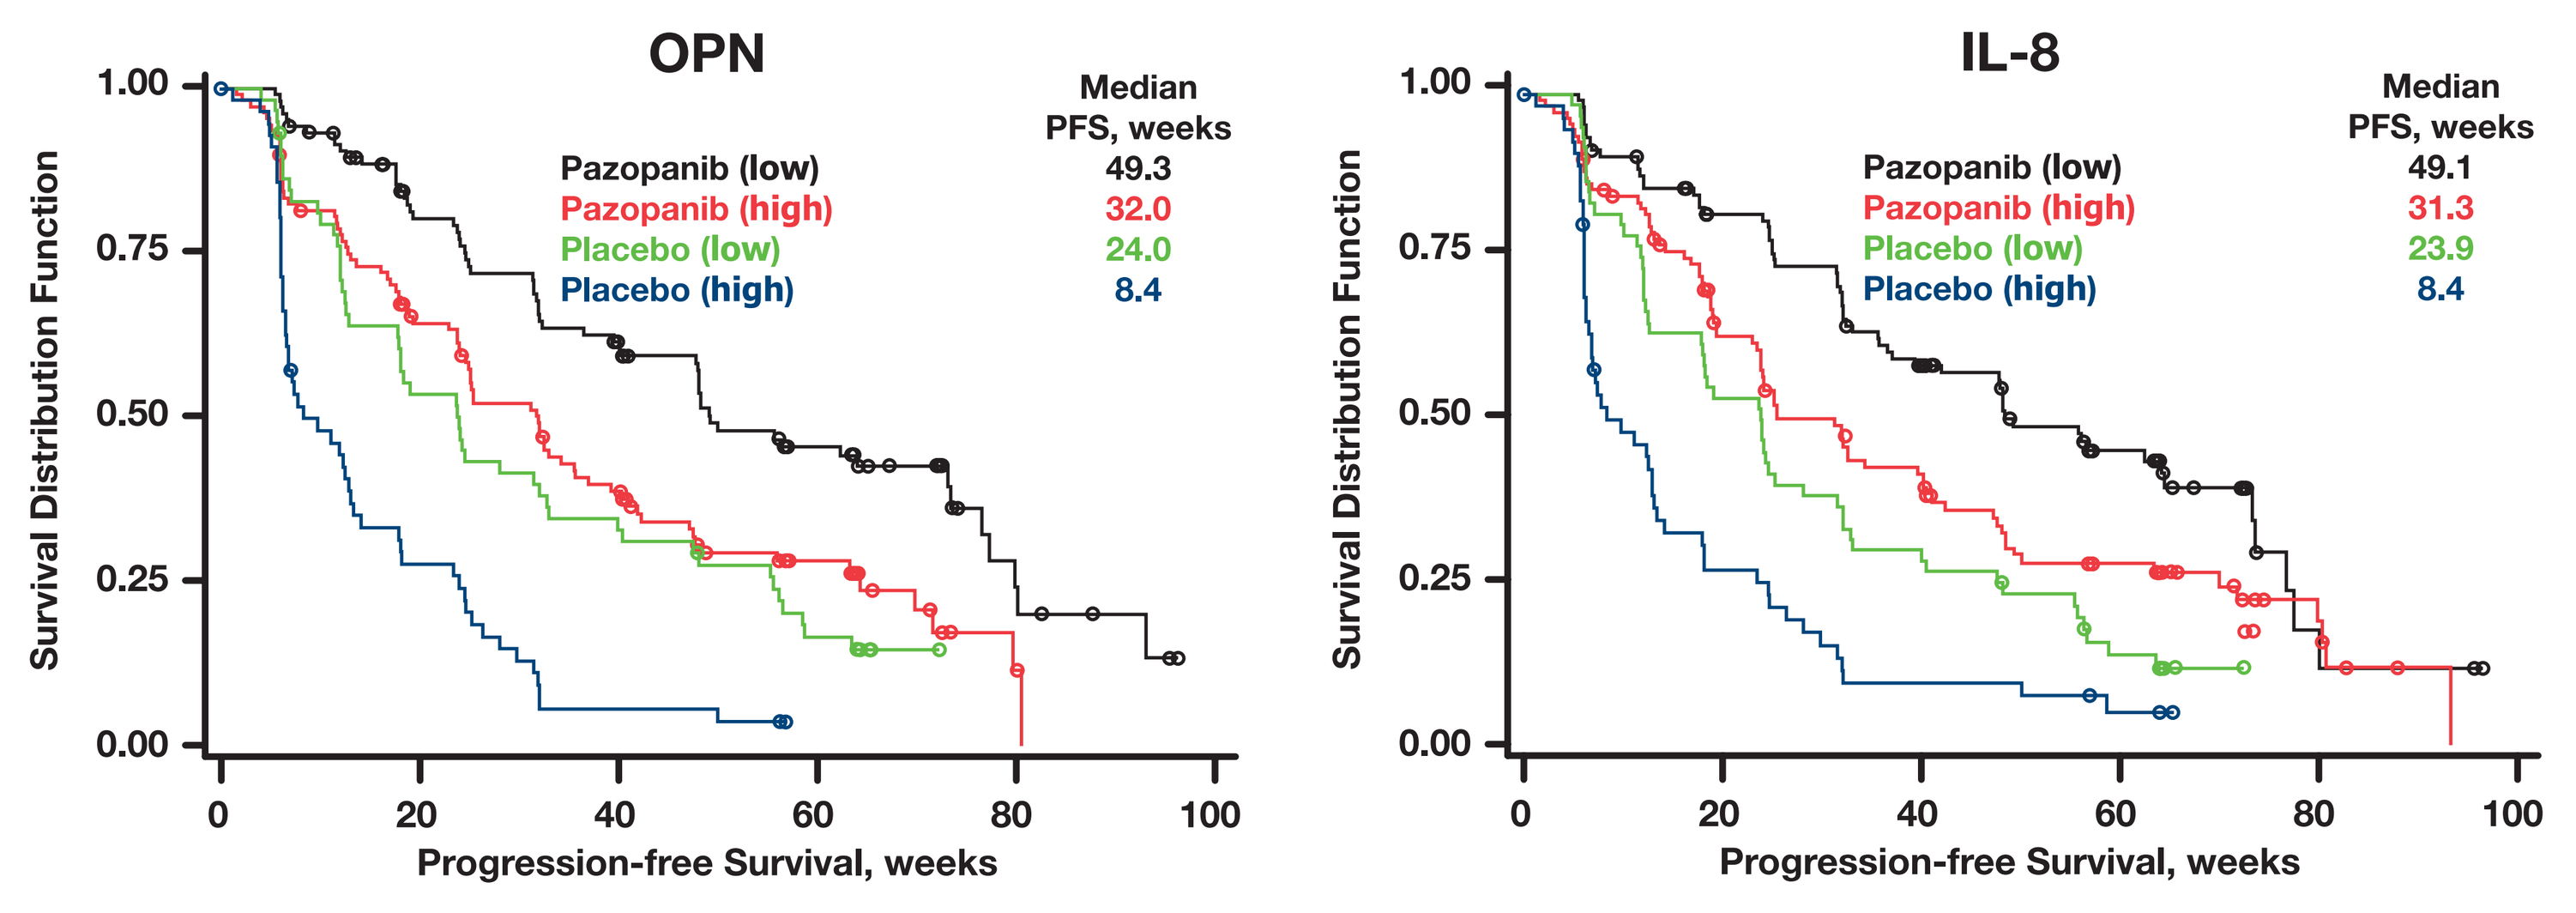
OPN
Median PFS, weeks
Pazopanib (low) 49.3
Pazopanib (high) 32.0
Placebo (low) 24.0
Placebo (high) 8.4
IL-8
Median PFS, weeks
Pazopanib (low) 49.1
Pazopanib (high) 31.3
Placebo (low) 23.9
Placebo (high) 8.4
Survival Distribution Function
Progression-free Survival, weeks

OPN Group	Median PFS, weeks: Pazopanib	Median PFS, weeks: Placebo	HR	95% CI	*P* (Pazopanib)	*P* (Placebo)	*P* (Interaction)
Low	49.3	24.0	0.43	0.29 - 0.64	.0004	< .0001	.3433
High	32.0	8.4	0.35	0.24 - 0.51			
Overall	39.6	13.4	0.42	0.33 - 0.55			

IL-8 Group	Median PFS, weeks: Pazopanib	Median PFS, weeks: Placebo	HR	95% CI	*P* (Pazopanib)	*P* (Placebo)	*P* (Interaction)
Low	49.1	23.9	0.41	0.28 -0.60	.0061	.0019	.4722
High	31.3	8.4	0.39	0.27 - 0.56			
Overall	39.6	13.4	0.42	0.33 - 0.55			

Predictive Markers

- Only IL-6 was predictive of PFS benefit
- The predictive value of baseline IL-6 levels was retained after correcting for ECOG performance status by covariate analysis

Figure 4. IL-6 PFS Analysis Using Baseline Median Cutoff

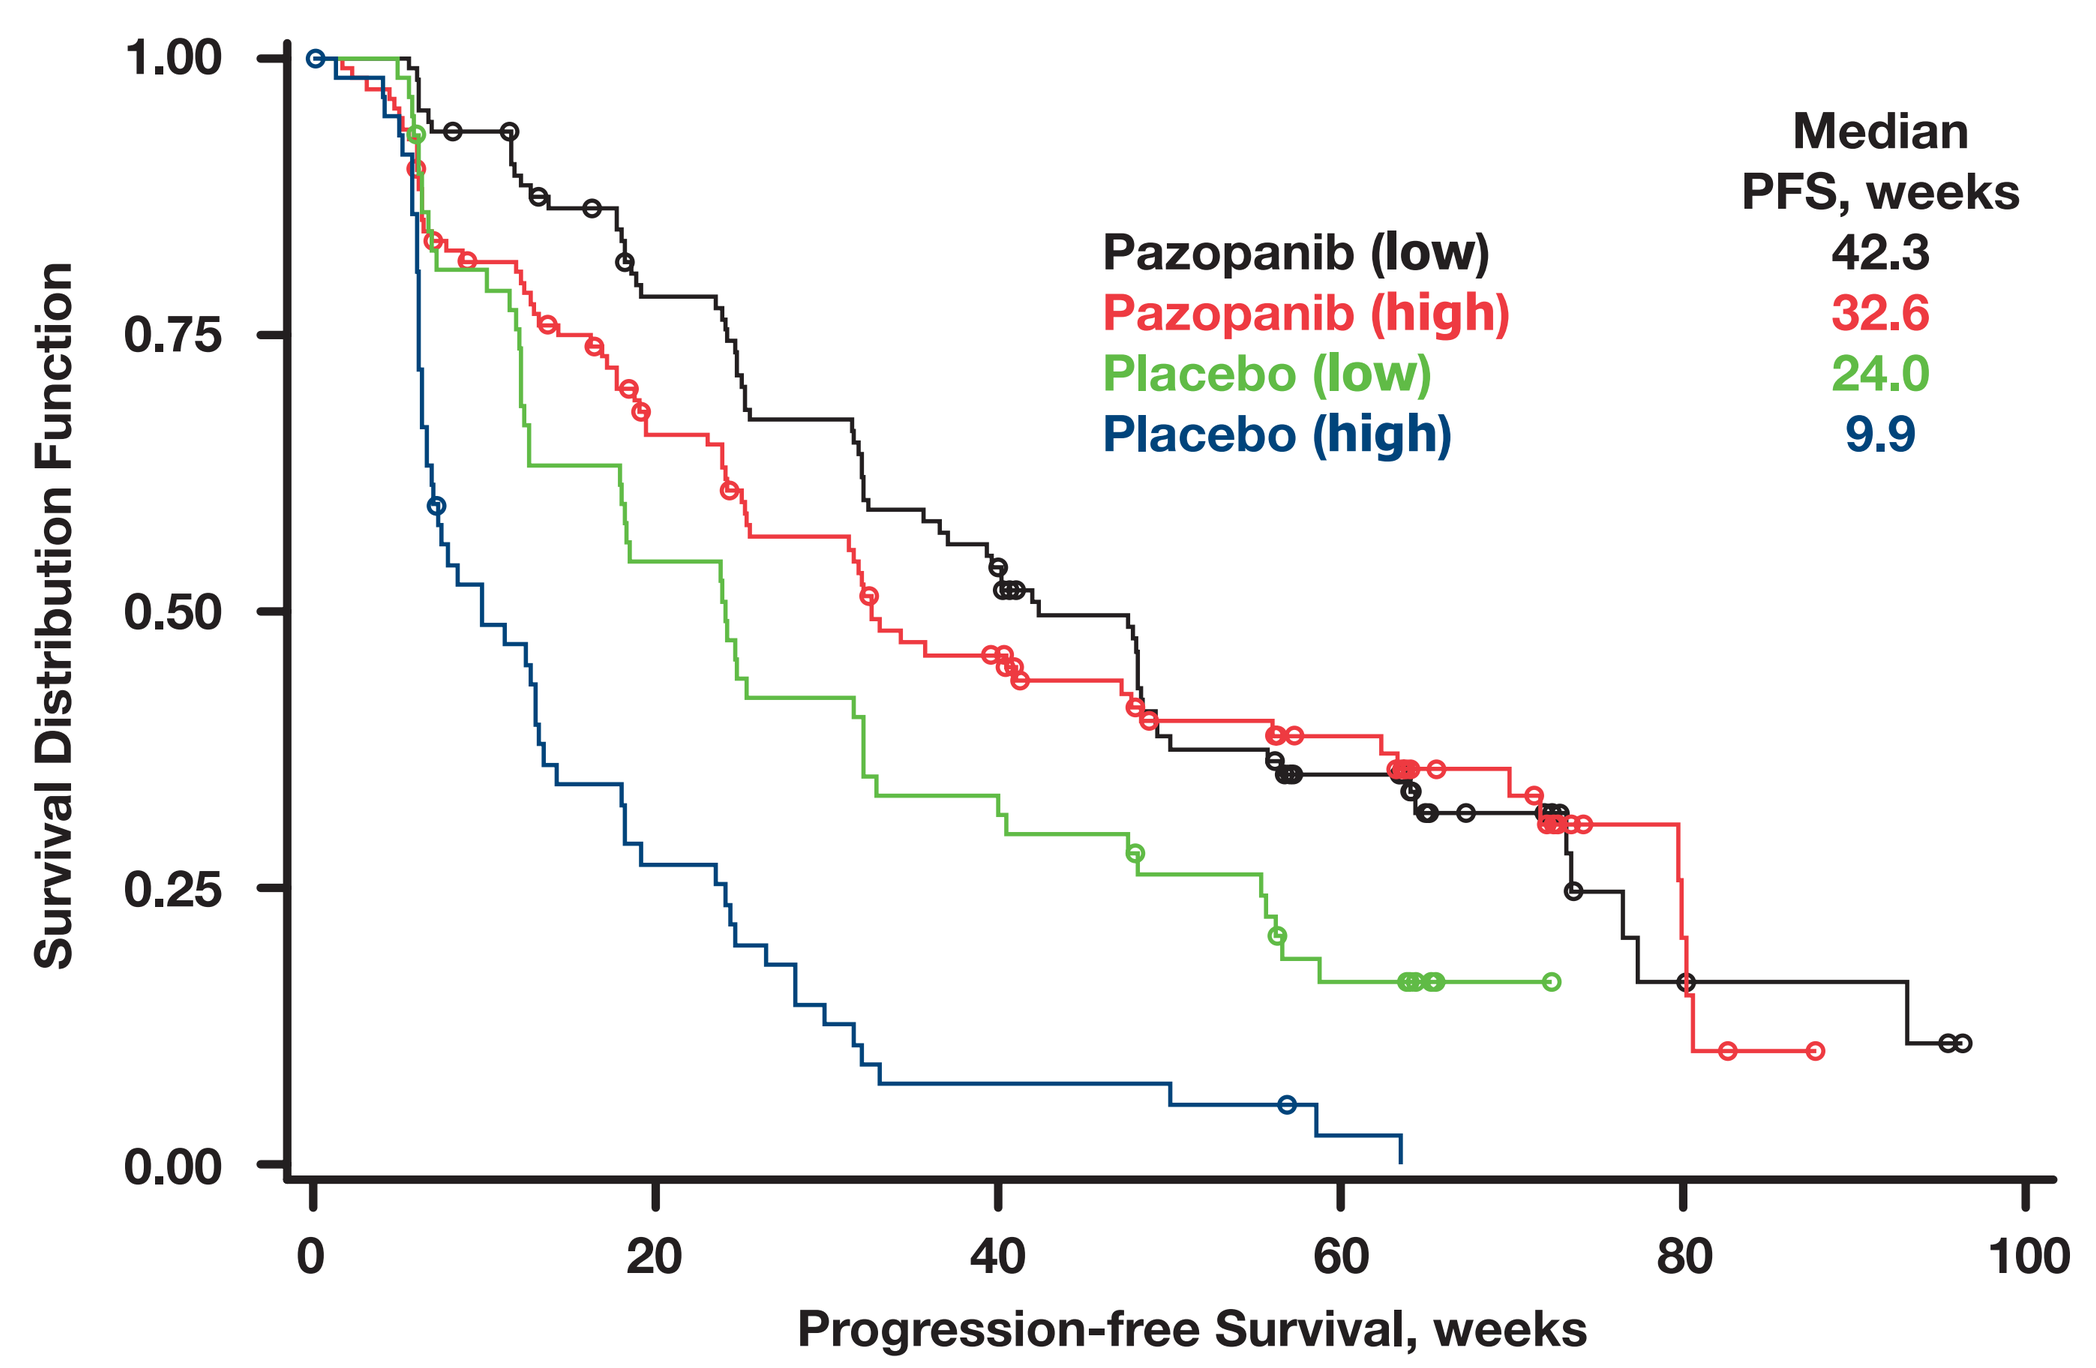
Median PFS, weeks
Pazopanib (low) 42.3
Pazopanib (high) 32.6
Placebo (low) 24.0
Placebo (high) 9.9
Survival Distribution Function
Progression-free Survival, weeks

IL-6 Group	Median PFS, weeks: Pazopanib	Median PFS, weeks: Placebo	HR	95% CI	*P* (Pazopanib)	*P* (Placebo)	*P* (Interaction)
Low	42.3	24.0	0.56	0.38 - 0.81	.445	< .001	.0089
High	32.6	9.9	0.32	0.21 - 0.44			
Total	39.6	13.4	0.42	0.33 - 0.55			

RESULTS (continued)

- In a phase II study, E-Selectin was also identified, and higher E-Selectin correlated with prolonged PFS
 - 2-factor analysis of the phase II study was confirmed in this phase III study

Figure 5. Two-Factor CAF (IL-6/E-Selectin) Signature PFS Analysis

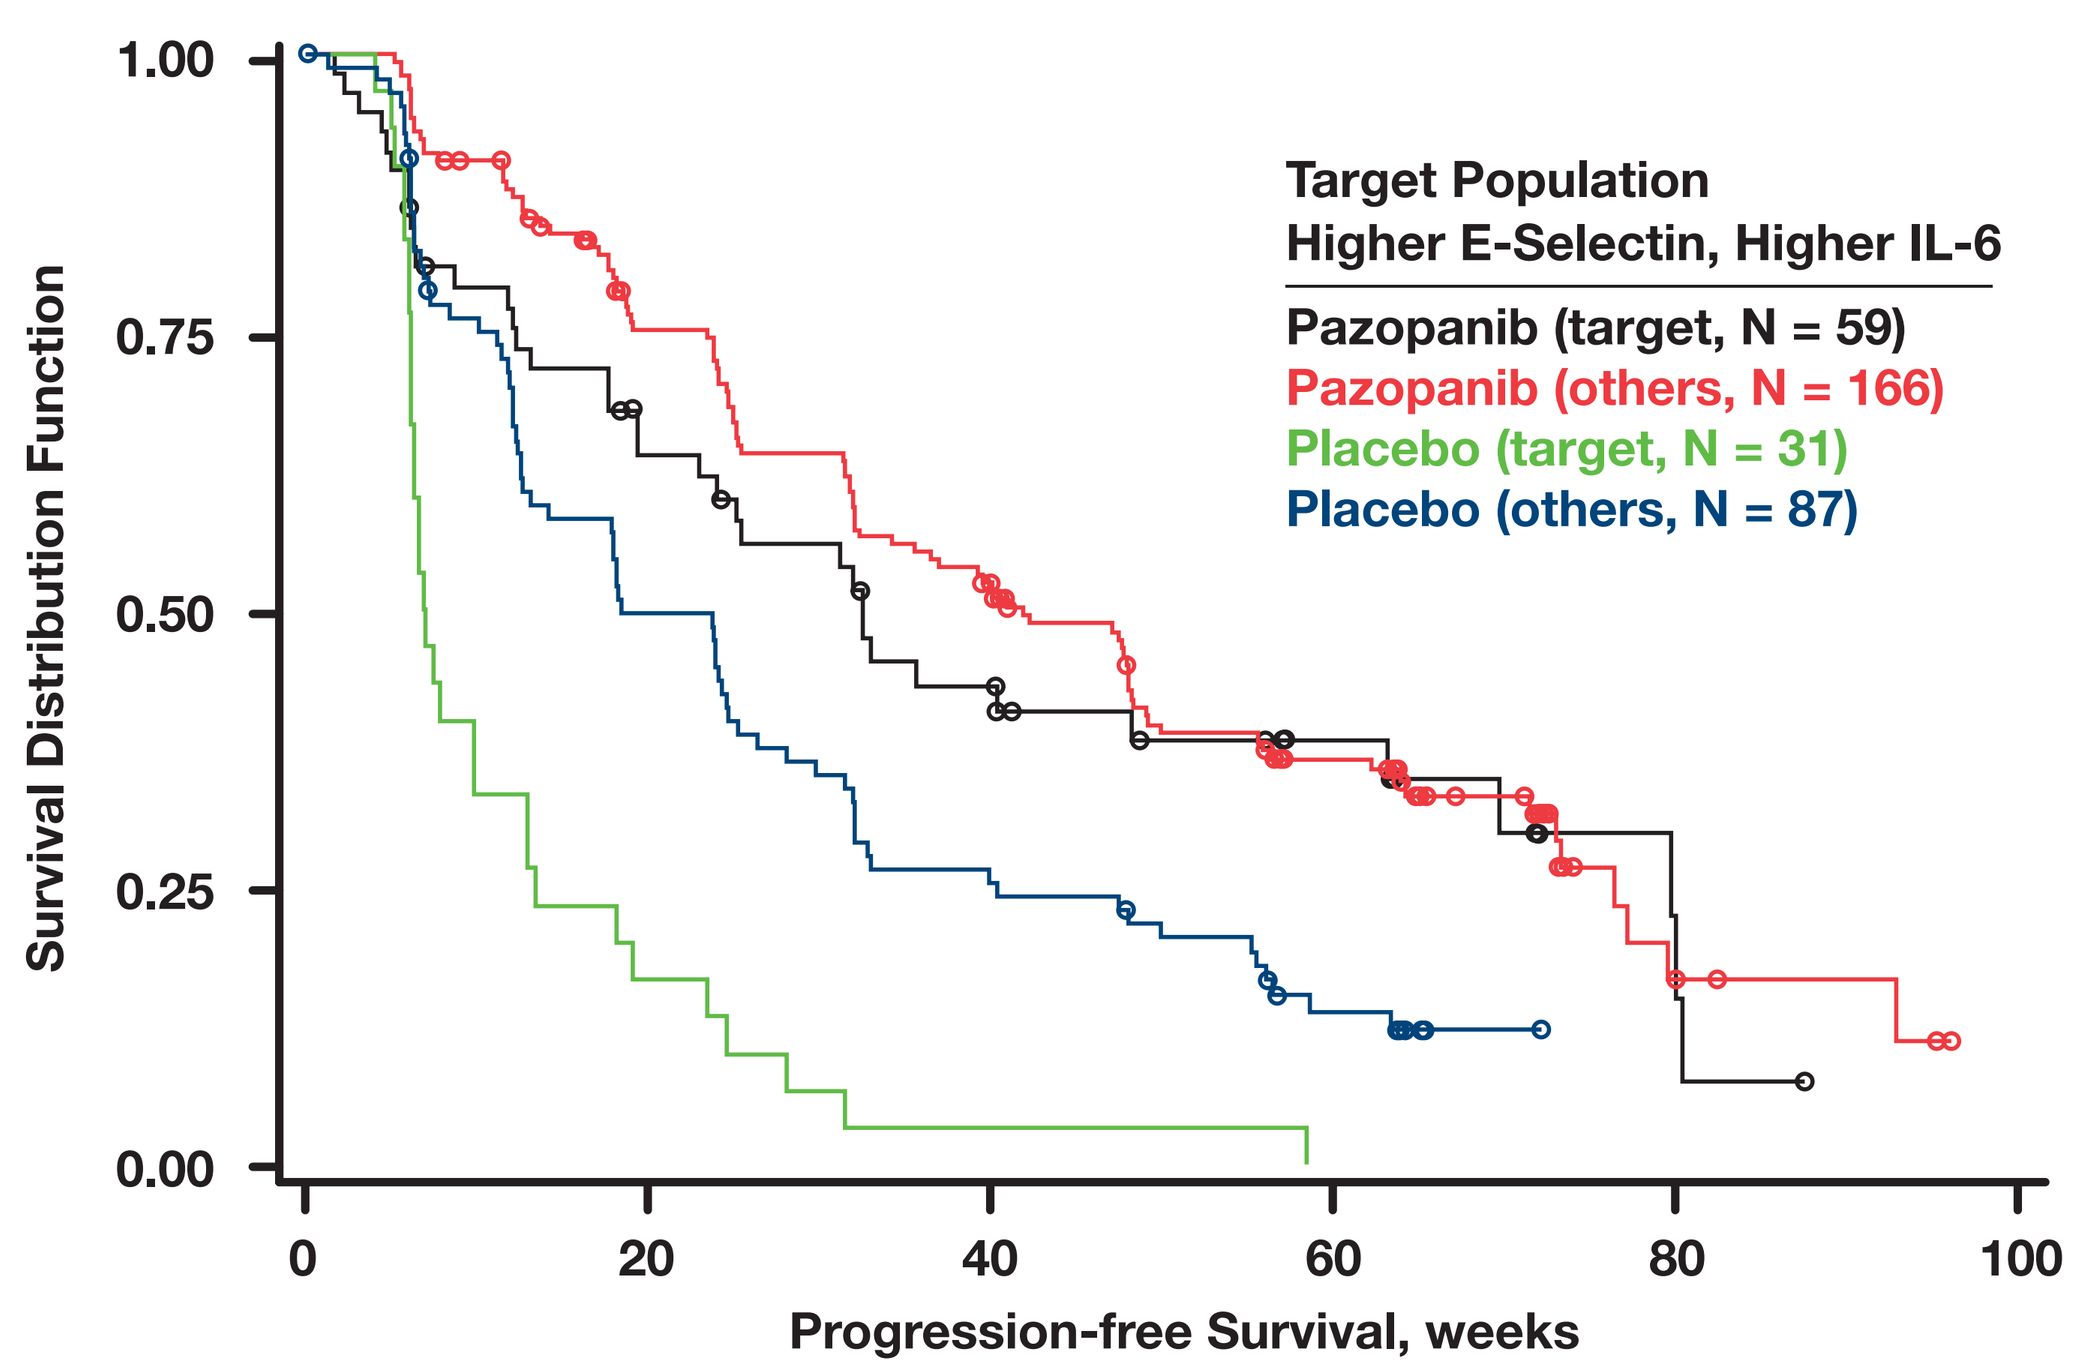
Target Population
Higher E-Selectin, Higher IL-6
Pazopanib (target, N = 59)
Pazopanib (others, N = 166)
Placebo (target, N = 31)
Placebo (others, N = 87)
Survival Distribution Function
Progression-free Survival, weeks

Group	Median PFS, weeks: Pazopanib	Median PFS, weeks: Placebo	HR	95% CI	*P* (Pazopanib)	*P* (Placebo)	*P* (Interaction)
Target	32.6	6.9	0.26	0.15 – 0.44	.653	< .001	.006
Others	42.0	18.4	0.48	0.36 – 0.66			
Total	39.6	13.4	0.42	0.33 – 0.55			

CONCLUSIONS

- Using CLIA-certified Searchlight multiplex platform, baseline levels of 7 candidate CAFs previously found to be associated with clinical outcomes in mRCC patients treated with pazopanib (HGF, IL-6, IL-8, E-Selectin, OPN, TIMP-1, VEGF) were tested in patients with available plasma samples (N = 344) enrolled in a randomized, placebo-controlled phase III clinical trial
- The association of 4 CAF markers with clinical benefit (PFS) were validated
 - IL-8 ($P < .006$), HGF ($P < .01$), OPN ($P < .001$), and TIMP-1 ($P < .006$)
- Analysis of data from the placebo arm showed the following CAF levels to be significantly associated with disease outcome (ie, prognostic): IL-6 ($P < .001$), IL-8 ($P < .002$), OPN ($P < .001$)
 - TIMP-1 showed a trend as a prognostic marker
- Baseline IL-6 levels were prognostic and predictive, with significantly different degrees of benefit observed in the low and high IL-6 groups

REFERENCES

1. Lam JS, et al. *World J Urol*. 2005;23:202-212.
2. Bellmunt J, Guix M. *BJU Int*. 2009;103:572-577.
3. Sonpavde G, Hutson TE. *Curr Oncol Rep*. 2007;9:115-119.
4. Australian Government Department of Health and Ageing. http://www.tga.gov.au/docs/html/orphand2.htm#p.
5. Davis J. http://www.medconnect.com.au/tabid/84/s2/Endocrinology/ct1/c337246/Pazopanib-Wins-Conditional-EU-Approval-for-Renal-Cell-Carcinoma/Default.aspx.
6. Instituto de Salud Publica de Chile. http://200.68.11.21/RegistrosISP/fiFichaProducto.asp?RegistroISP=F-18018/10.
7. Kidney Cancer Canada. http://www.kidneycancercanada.org/main.php?p=600&lan=1.
8. U.S. Department of Health and Human Services Food & Drug Administration. (http://www.fda.gov/AboutFDA/CentersOffices/CDER/ucm187509.htm).
9. Tran HT, et al. *J Clin Oncol*. 2010;28:15s (abstract 4522).

ACKNOWLEDGMENTS

This study was sponsored by GlaxoSmithKline, Philadelphia, Pennsylvania.

Appreciation is expressed to the patients and their families, as well as the many investigators who participated in the VEG102919 study. We also thank Aushon Biosystems, the GSK VEG102919 clinical trial team, and Robert Gagnon of GSK.

Presented at: 2011 Genitourinary Cancers Symposium (ASCO GU); February 17-19, 2011; Orlando, Florida.